ATOPIC ECZEMA SCORE OF EMOTIONAL CONSEQUENCES (AESEC): AN INSTRUMENT TO MEASURE EMOTIONAL CONSEQUENCES IN ADULTS

Antje Finkwagner (1), Bernd W.M. Arents (1)*, Giuseppe De Carlo (1), Ute Mensing (2), Irmtraud A. Seitz (2), Johannes Ring (3)
1 European Federation of Allergy and Airway Diseases Patient's Associations, Brussels, Belgium;
2 IMAS International, Munich, Germany
3. Department of Dermatology and Allergy, Biederstein, TU Munich, Germany
* Patient with severe atopic eczema

European Federation of Allergy and Airways Diseases Patients' Associations

BACKGROUND

Atopic eczema (AE) is a common inflammatory skin disease affecting 1-3% of the adults in the EU. Symptoms of AE include dry, patchy skin lesions and intense itching. It's remitting and relapsing in nature and can have huge consequences for physical and psychological wellbeing. For a better understanding of the impact of Atopic Eczema on patients quality of life we identified the need to develop a questionnaire that could assess the emotional consequences of suffering from Atopic Eczema.

OBJECTIVE

1) To develop and validate a questionnaire to evaluate and measure emotional consequences of suffering from AE.

2) To use the questionnaire in a survey with n= 1189 patients with severe AE in 9 European countries.

METHODS

An extensive item list was compiled based on previous experience with QoL studies. Additionally, through social media people with AE (n= 235) were asked how AE was impacting them in daily life, and those items were aligned with the initial list. The items (n=75) were intentionally reworded into positive and negative statements to avoid internal negative bias, and checked by specialists in psychology, research and language. We investigated how the patients see their own situation with others (external situation) and how they are able to cope with their disease. A 4-point Likert scale was selected (applies fully, somewhat applies, rather not, does not apply at all). A questionnaire containing 37 items was pretested online in 5 EU-countries (n=100) to select the most significant and reliable statements. 28 items were chosen based on reliability and inter-item correlation. AESEC scores are: 0-27 no/small impact on life; 28-39 moderate impact; 40-52 large impact; 53-84 very large impact on life. The questionnaire was then embedded in a larger one, which included the validated instruments POEM, HADS-D7 and DLQI. After approval of local Ethics Committees and (mainly) recruitment via physicians, adults with severe AE in nine EU-countries were interviewed by telephone.

RESULTS

The reliability of the 28-items questionnaire is high (α=0.900) with low inter-item correlation (ρ=0.208). Reliability based on three psychological dimensions is high (α=0.800-0.822) with weak inter-item correlation (ρ=0.278-0.324). When using a three-factor analysis the reliability is high (α=0.802-0.882) with a weak/moderate inter-item correlation (ρ=0.294-0.428). Correlation with HADSD7 is 0.377, 0.542 with DLQI, 0.449 with POEM and 0.440 with self-reported severity. Based on score-distribution/standard deviation, HADS-D7, POEM and DLQI, the AESEC scores (0-84) can be interpreted as: 0-27=small, 28-39=moderate, 40-52=large and ≥53=very large. Applied to the participants in this EU-study, 43% experience small, 27% moderate, 21% large and 9% very large consequences.
Generally, patients agree in most cases on **envying people with normal skin** and that **itching drives crazy**. Additionally, the **desire to hide the eczema** and **sadness** about having AE can also be noticed. Furthermore there are clear tendencies for **feeling guilty** about scratching, **avoidance** of physical contact, **embarrassment** regards the skin appearance and **being angered** by the disease, especially in people who are in a moderate to severe condition.

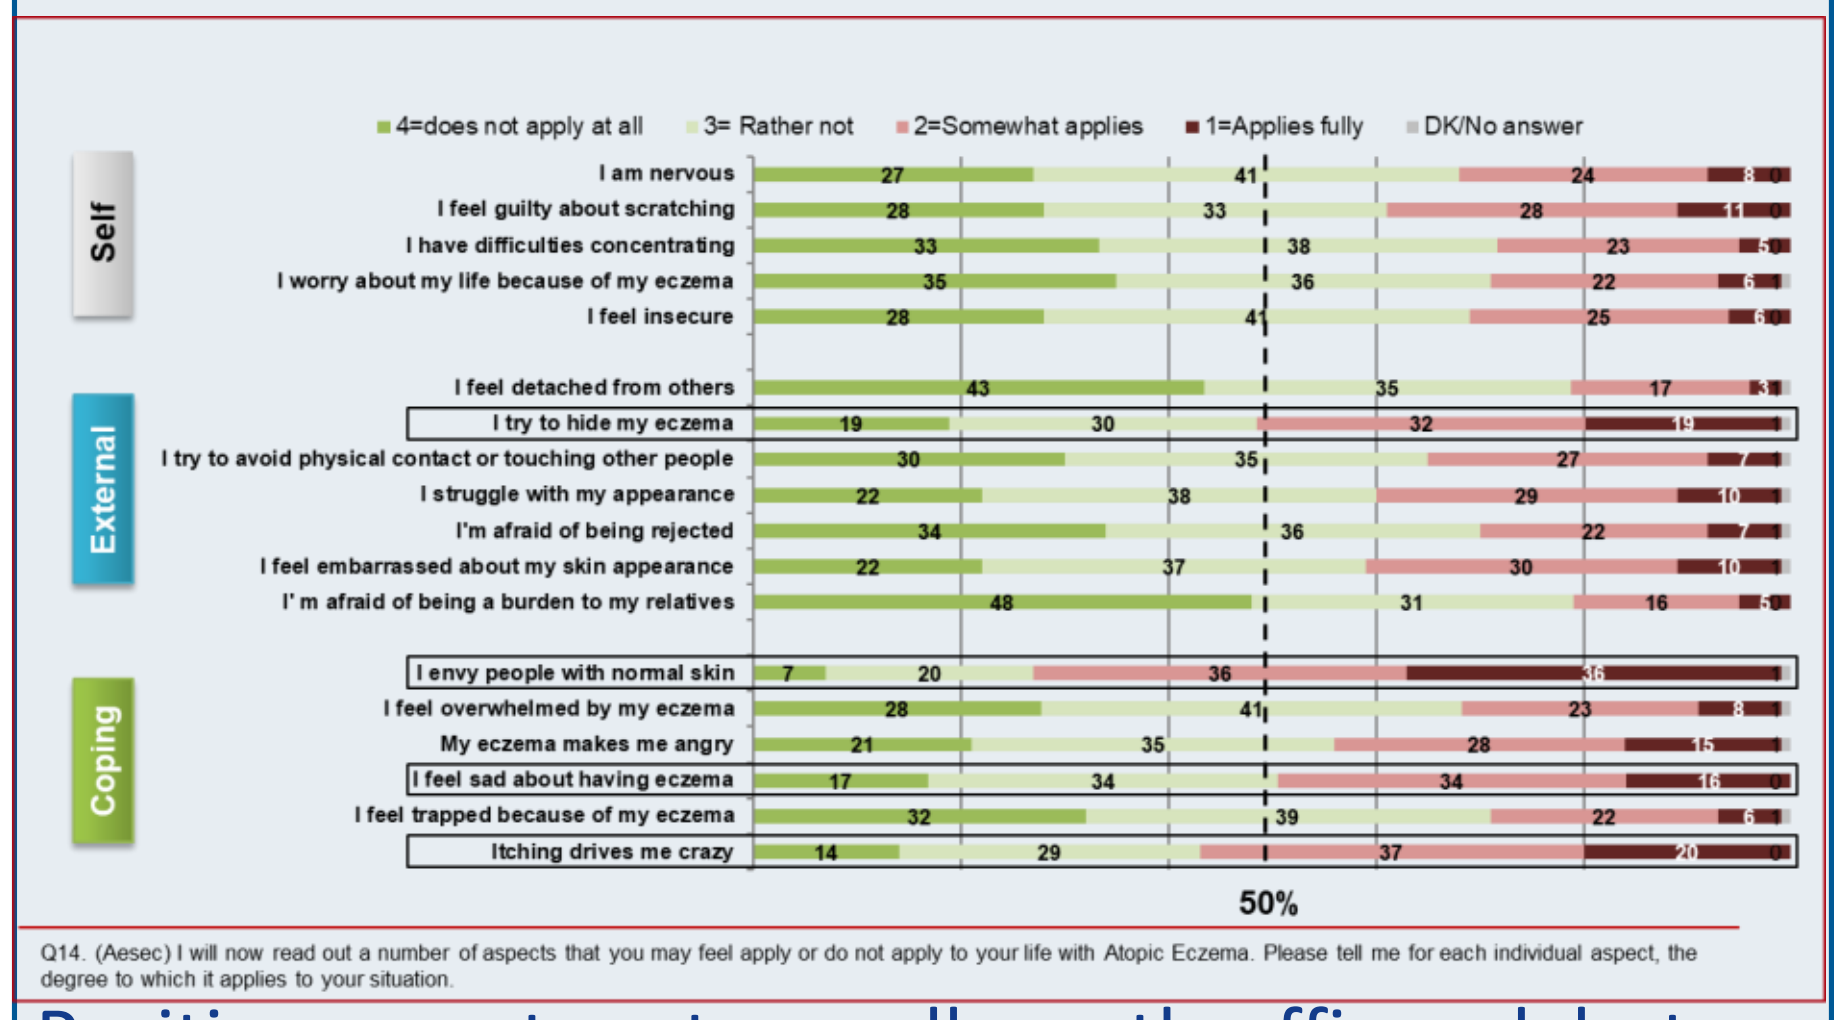
4=does not apply at all
3= Rather not
2=Somewhat applies
1=Applies fully
DK/No answer
Self
I am nervous
I feel guilty about scratching
I have difficulties concentrating
I worry about my life because of my eczema
I feel insecure
External
I feel detached from others
I try to hide my eczema
I try to avoid physical contact or touching other people
I struggle with my appearance
I'm afraid of being rejected
I feel embarrassed about my skin appearance
I'm afraid of being a burden to my relatives
Coping
I envy people with normal skin
I feel overwhelmed by my eczema
My eczema makes me angry
I feel sad about having eczema
I feel trapped because of my eczema
Itching drives me crazy
50%

Q14. (Aesec) I will now read out a number of aspects that you may feel apply or do not apply to your life with Atopic Eczema. Please tell me for each individual aspect, the degree to which it applies to your situation.

Positive aspects get overall mostly affirmed, but the level of severity affects the level of affirmation and plays a major role. Patients that are currently affected (especially severe or very severe) show **less optimism, less balance, feel less in control** of the eczema and also show increased problems with **intimacy**.

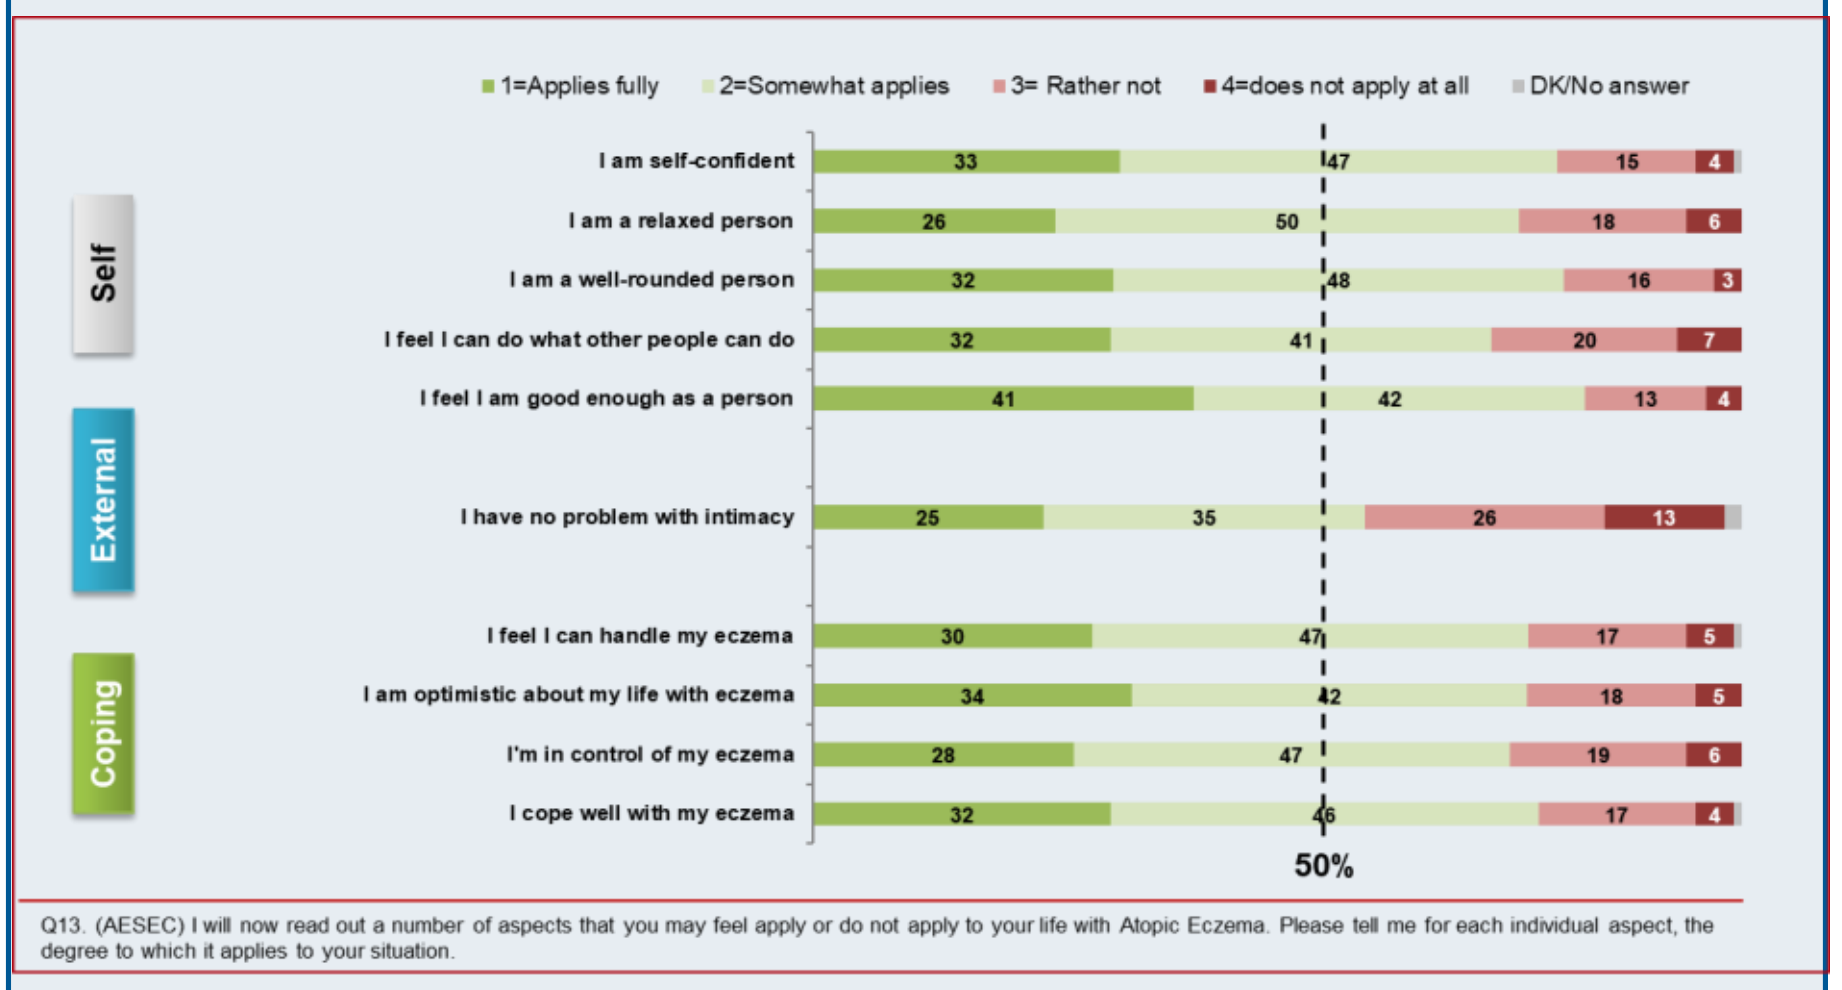
1=Applies fully
2=Somewhat applies
3= Rather not
4=does not apply at all
DK/No answer
Self
I am self-confident
I am a relaxed person
I am a well-rounded person
I feel I can do what other people can do
I feel I am good enough as a person
External
I have no problem with intimacy
Coping
I feel I can handle my eczema
I am optimistic about my life with eczema
I'm in control of my eczema
I cope well with my eczema
50%

Q13. (AESEC) I will now read out a number of aspects that you may feel apply or do not apply to your life with Atopic Eczema. Please tell me for each individual aspect, the degree to which it applies to your situation.

When looking at the POEM sub-groups, it is clear that increased severity leads to an increased mean AESEC score. Patients, which are grouped as severe (POEM) show a mean of 43 and "very severe" cases reach 47.

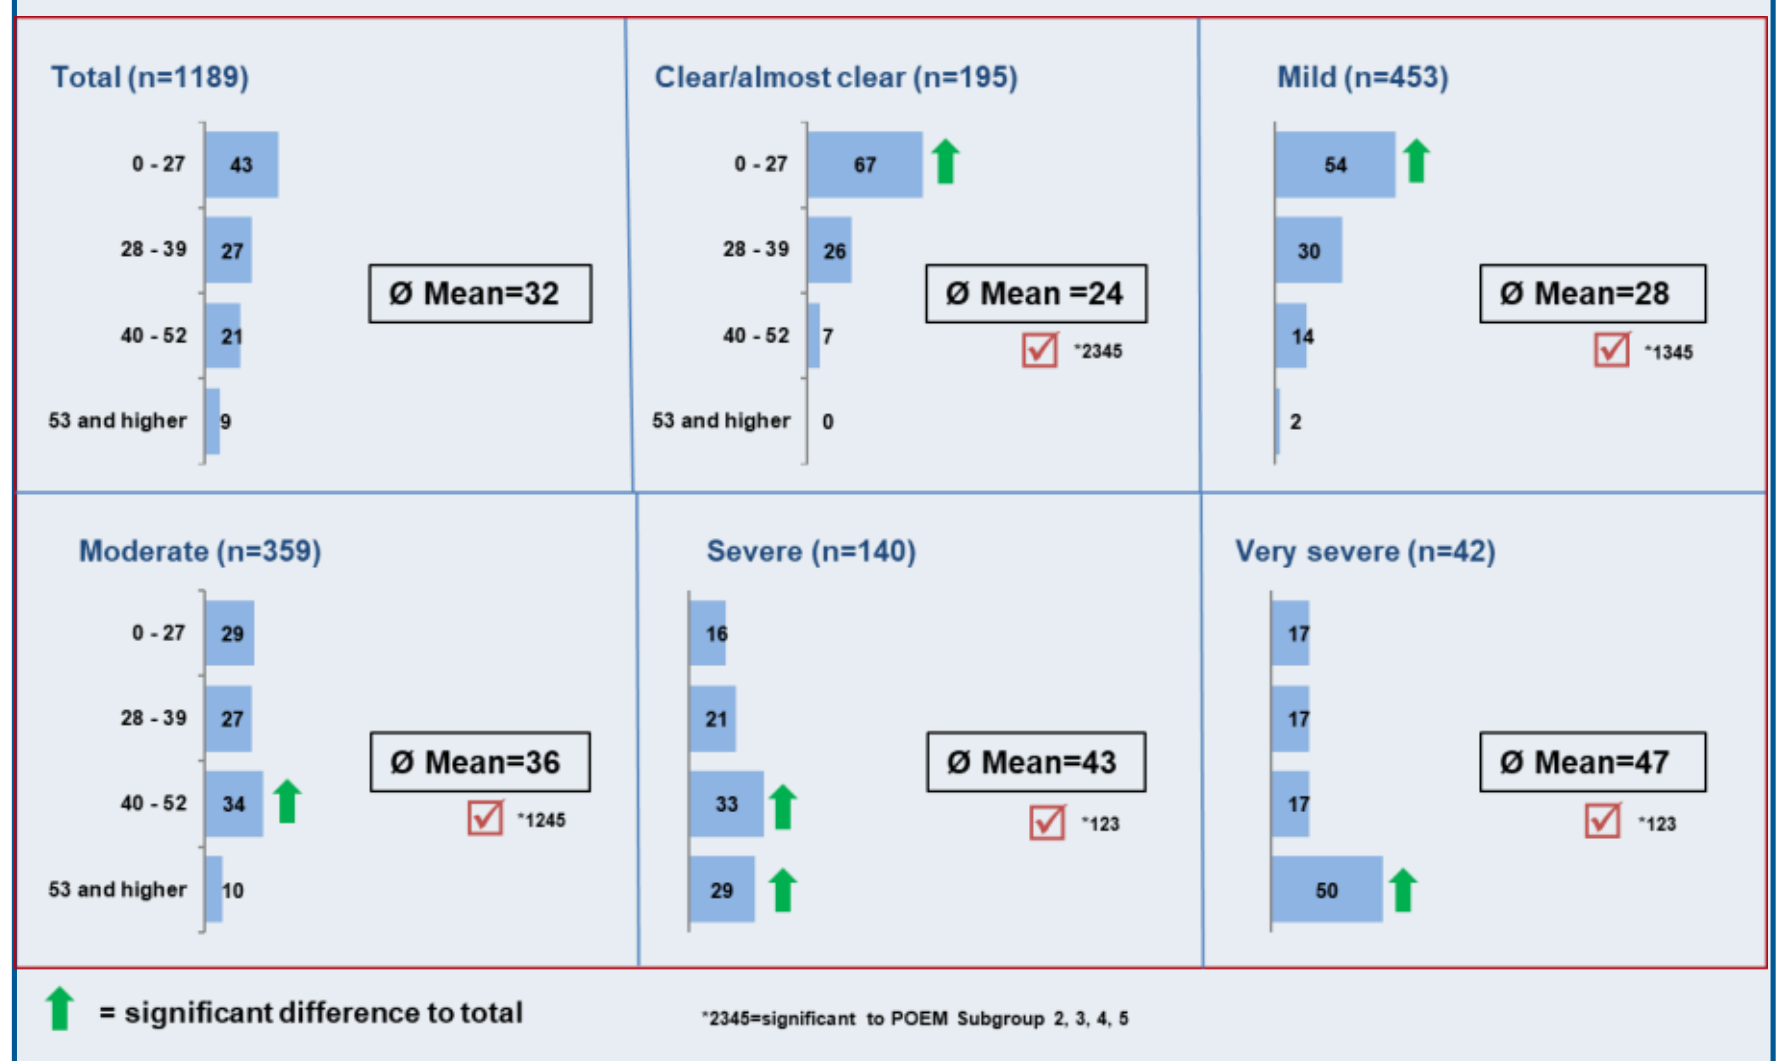
Total (n=1189)
Ø Mean=32
Clear/almost clear (n=195)
Ø Mean =24
Mild (n=453)
Ø Mean=28
Moderate (n=359)
Ø Mean=36
Severe (n=140)
Ø Mean=43
Very severe (n=42)
Ø Mean=47
0 - 27
28 - 39
40 - 52
53 and higher
= significant difference to total
*2345=significant to POEM Subgroup 2, 3, 4, 5

Also, when considering the self-assessed severity level, a clear increase of the AESEC scores can be noticed - from 24.40 amongst people with an almost clear skin to 46.48 amongst severe sufferers. This confirms a considerable tendency: **from a low impact on the life quality of patients which are currently fine to a high/very high impact for patients that currently show a severe condition.**

(1) (Almost) clear (n=269) - self-reported (=100%)					
AESEC Mean	SD (+/-)	Score 1: 0-27 „no/small"	Sore 2: 28-39 „moderate"	Score 3 40-52 „large"	Score 4: 53 ++ „very large"
24,40	9,222	**72%**	**22%**	**6%**	**0%**

(2) Mild (n=371) - self-reported (=100%)					
AESEC Mean	SD (+/-)	Score 1: 0-27 „no/small"	Sore 2: 28-39 „moderate"	Score 3 40-52 „large"	Score 4: 53 ++ „very large"
29,37	12,726	**46%**	**32%**	**18%**	**4%**

(3) Moderate (n=434) - self-reported (=100%)					
AESEC Mean	SD (+/-)	Score 1: 0-27 „no/small"	Sore 2: 28-39 „moderate"	Score 3 40-52 „large"	Score 4: 53 ++ „very large"
35,73	13,574	**31%**	**27%**	**32%**	**10%**

(4) Severe (n=115) - self-reported (=100%)					
AESEC Mean	SD (+/-)	Score 1: 0-27 „no/small"	Sore 2: 28-39 „moderate"	Score 3 40-52 „large"	Score 4: 53 ++ „very large"
46,45	14,652	**10%**	**23%**	**29%**	**38%**

Emotional impact correlates with current severity

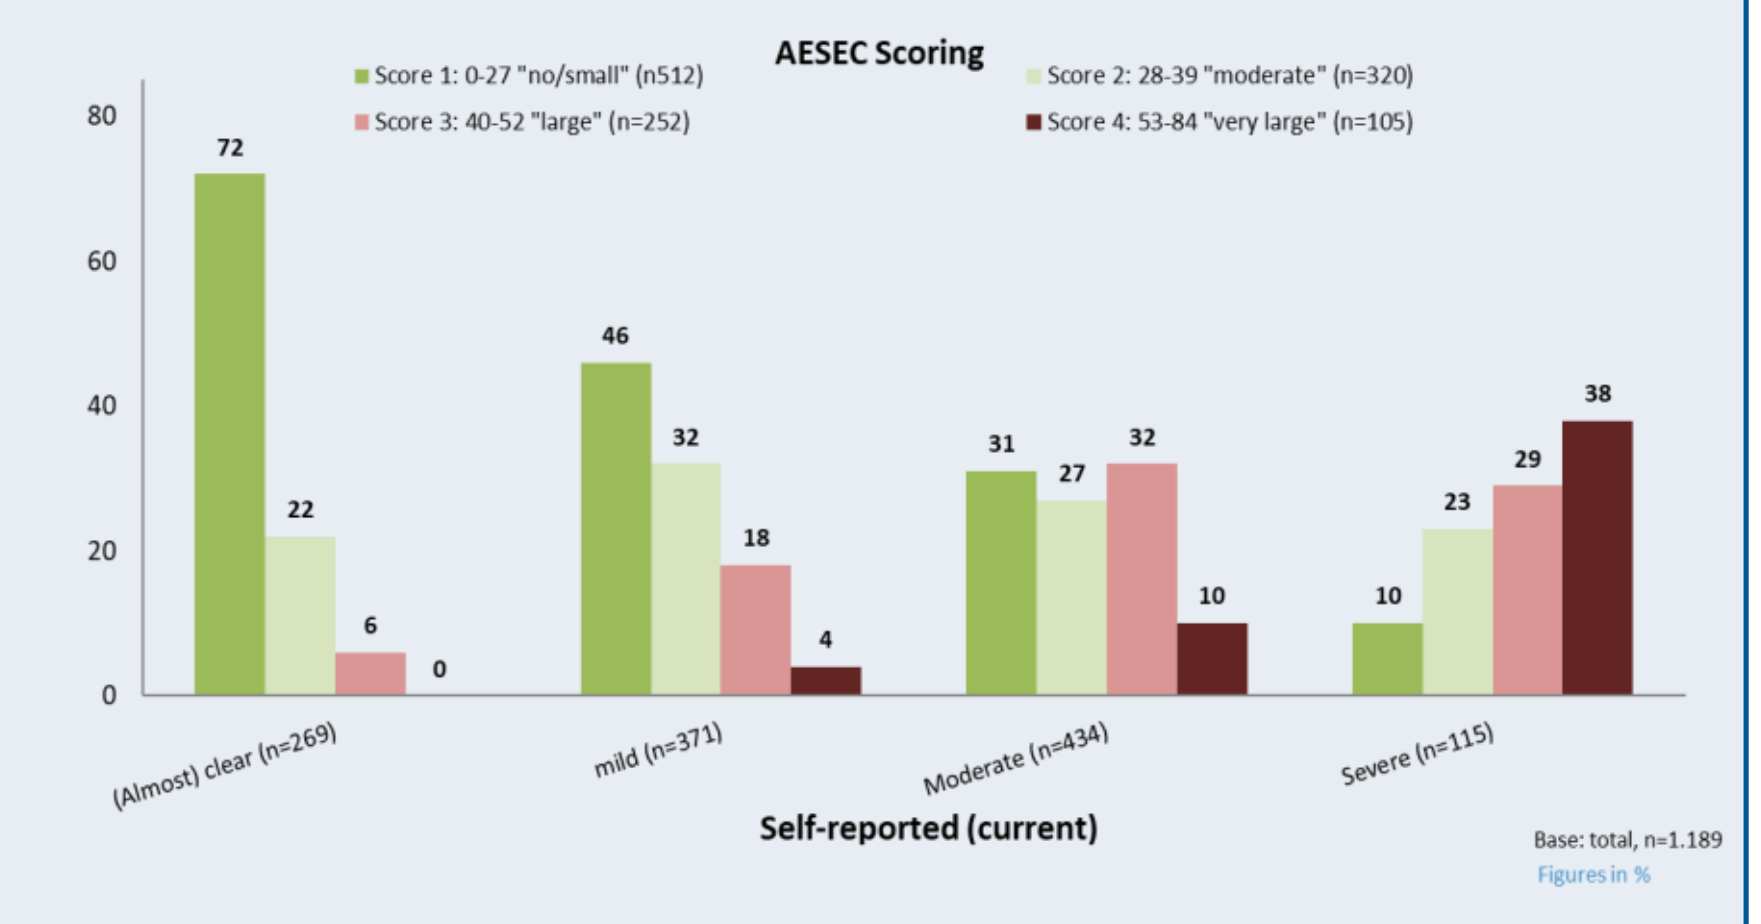
AESEC Scoring
Score 1: 0-27 "no/small" (n512)
Score 2: 28-39 "moderate" (n=320)
Score 3: 40-52 "large" (n=252)
Score 4: 53-84 "very large" (n=105)
(Almost) clear (n=269)
mild (n=371)
Moderate (n=434)
Severe (n=115)
Self-reported (current)
Base: total, n=1.189
Figures in %

CONCLUSIONS

This study shows that the Atopic Eczema Score of Emotional Consequences (AESEC) is a valid instrument to measure emotional consequences of having severe AE as an adult, with 30% of those patients experiencing (very) large consequences.

This real-life study confirms-that adults with currently a more severe form of AE are **suffering more than would be deemed acceptable**.

In particular more efforts are needed in (i) Improving the **acceptance and understanding** of the disease among the public, health care professionals, national and EU policy makers, and health care payers (e.g. insurers) (ii) Providing **access to better and affordable health care** for the patients, with higher financial support from health insurances (iii) **Investing in research** for the development of more effective treatments (iv) Relieving the **psychological and emotional** burden of this disease.

ACKNOWLEDGEMENT

EFA wishes to thank the sustainable funding partners Sanofi Genzyme and Regeneron whose unrestricted educational grants made this project, and the creation of the report possible. We thank them for their kind support to assess the patients' quality of life and the burden of Atopic Eczema.